DEVELOPMENT OF A PERSONALIZED CELLULAR EX-VIVO CBL-B SILENCING CANCER IMMUNE THERAPY

Marc Oliver Salzberg[1)], Guenther Lametschwandtner[1)], Manfred Schuster[1)], Hans Loibner[1)], Monika Sachet[2)], Hubert Hayden[2)], Michaela Hassler[2)], Josef Friedl[2)], Pierre Triozzi[3)]

1) Apeiron-Biologics, Vienna, Austria 2) Department of Surgery, General Hospital of Vienna 3) Wake Forest University, Winston-Salem, NC

Background:

- Cbl-b is a Ring-Finger E3 Ubiquitin ligase
- Cbl-b is a key negative regulator of lymphocyte activation
- Loss of cbl-b uncouples T-cell activation from requirement for costimulation and overcomes suppressor effects of TGF-β

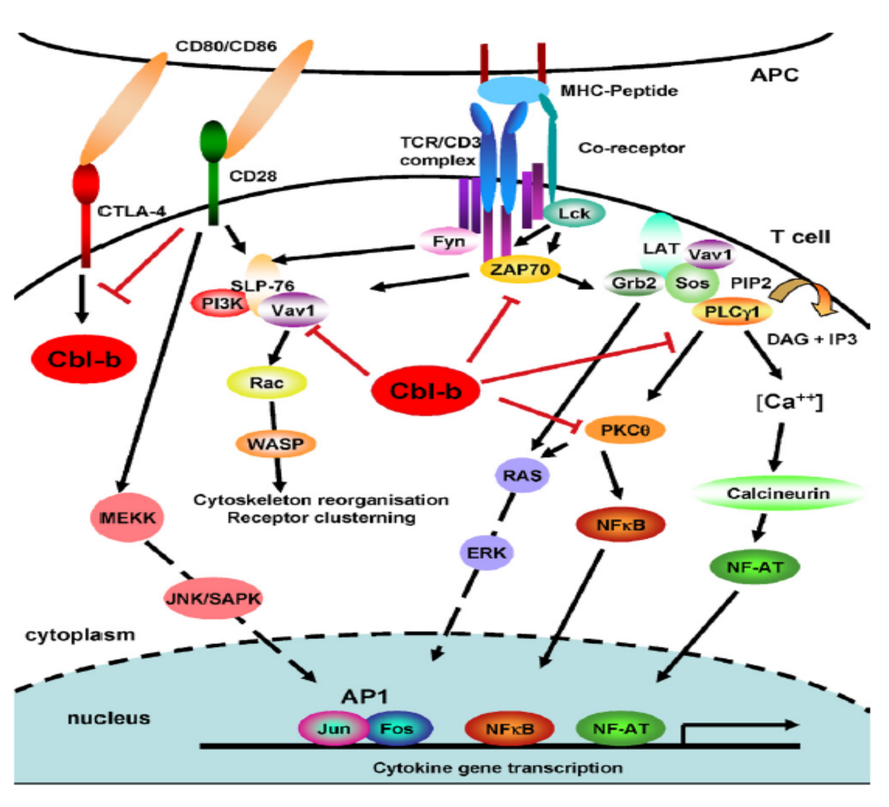
APC
T cell
Cbl-b
cytoplasm
nucleus
AP1
Cytokine gene transcription

Figure 1. The E3 Ubiquitin ligase cbl-b serves as a checkpoint to limit immune cell activation

Cbl-b modulates the activity of signaling pathways by binding/ubiquitinylating key molecules for signal transduction.

Fig. from Loeser & Penninger, Seminars in Immunology, 2007

- TCR stimulated cbl-b deficient T cells show enhanced proliferation and cytokine production (IFN-γ, IL-2 and TNF-α).
- Cbl-b deficient mice can mount efficient immune responses against weak immunogens

Target Validation in cbl-b deficient mice*

- Cbl-b knock-out mice spontaneously reject autologous tumors
- Adoptive cell transfer (ACT) of cbl-b deficient CD8 T cells is sufficient to eradicate already established tumors
- Cbl-b deficient T cells are less sensitive to suppression by T-regulatory cells
- Cbl-b deficient mice suffer only from limited autoimmunity and can reach normal lifespans

*(*Loeser et al., J. Exp Med 2007, Chiang et al., J. Clin Invest 2007)*

In vivo Target Validation of cbl-b Silencing*

- Adoptive Transfer of cbl-b silenced CD8 T cells together with DCs suppress tumor growth during treatment period (Fig. 2).

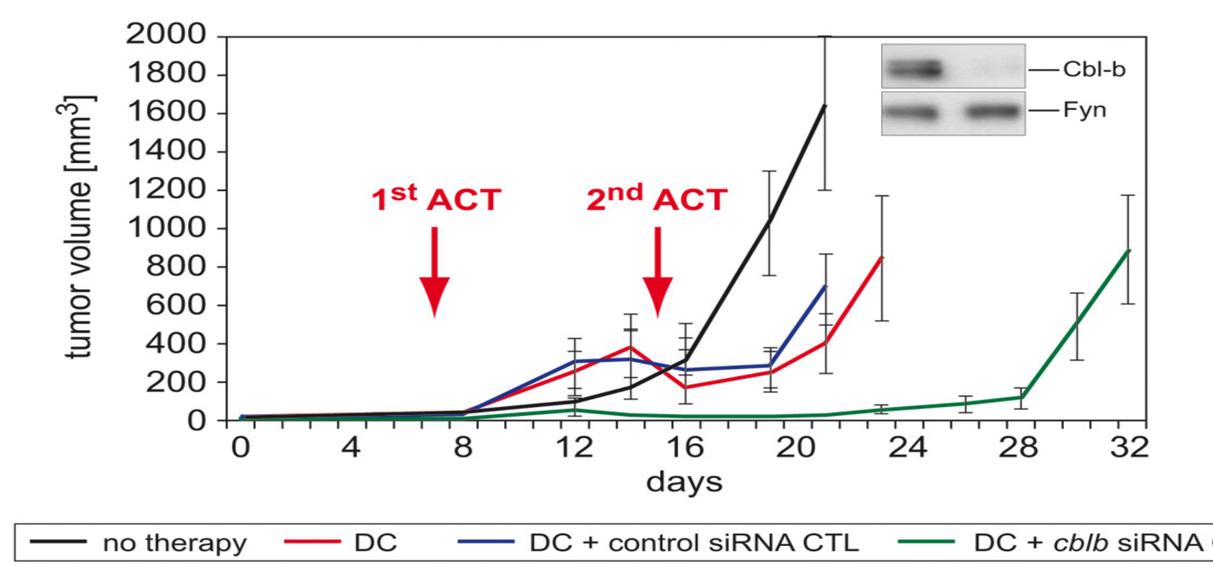
tumor volume [mm³]
1st ACT
2nd ACT
days
Cbl-b
Fyn
no therapy
DC
DC + control siRNA CTL
DC + cblb siRNA CTL

Figure 2. CD8 T cells were transfected with cbl-b siRNA or control and 3*10^6 cells were transferred to B16 tumor-harboring mice (1st & 2nd ACT), with prior vaccination of OVA-loaded DCs. As controls, either untreated mice or mice with DC-therapy alone are shown

**Hinterleitner et al., PLoS 2012*

- Cbl-b silenced CD8 T cells accumulate in the tumor and TILs isolated from the explanted tumor 5 days after cell transfer produce enhanced amounts of IL-2 and IFN-γ.

Target Validation in human T Cells

Cbl-b silencing in human T cells enhanced IL-2 and IFN-γ production, reproducing the phenotype of cbl-b deficient murine T cells and displayed more sustained activation following TCR stimulation (Fig. 3).

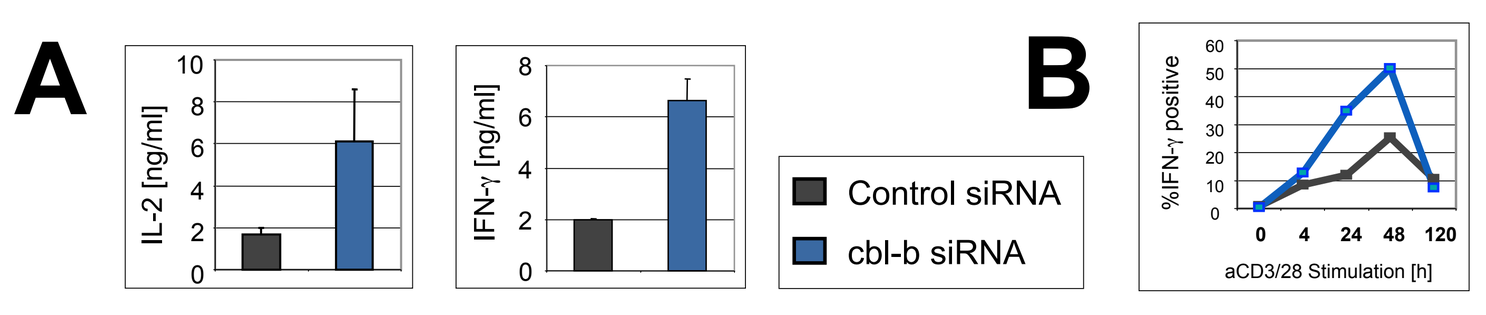
A
IL-2 [ng/ml]
IFN-γ [ng/ml]
Control siRNA
cbl-b siRNA
B
%IFN-γ positive
aCD3/28 Stimulation [h]

Figure 3. A) CD4 T cells were transfected with cbl-b or control siRNA (Dharmacon) and stimulated for 24h using aCD3/28 mAbs. IL-2 and IFN-γ production was determined by ELISA. **B)** T cells from A) were fixed at indicated time points following TCR stimulation and IFN-γ production was determined by icFACS.

Cbl-b silenced human T cells proliferated more vigorously and higher fractions of T cells completed more than 2 cell divisions following TCR stimulation. Moreover, cbl-b silenced T cells were less sensitive to suppression by TGF-β (Fig. 4).

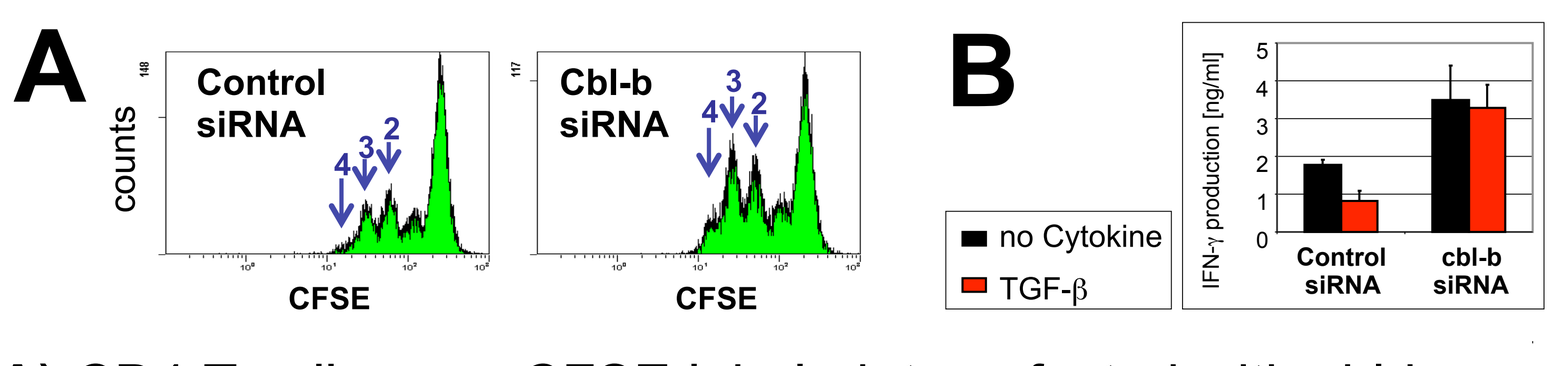
A
Control siRNA
Cbl-b siRNA
counts
CFSE
CFSE
B
no Cytokine
TGF-β
IFN-γ production [ng/ml]
Control siRNA
cbl-b siRNA

Figure 4. A) CD4 T cells were CFSE labeled, transfected with cbl-b or control siRNA and stimulated for 3 days using aCD3/28 mAbs. CFSE content of cells was determined by FACS. **B)** CD8 T cells were transfected with cbl-b or control siRNA and stimulated for 24h either in the absence or presence of 5ng/ml TGF-β. IFN-γ production was determined by ELISA

Cbl-b Silencing in human PBMCs

A transfection procedure for simultaneous and highly efficient transfection of all major leukocyte types present in human PBMCs was developed (Fig. 5).

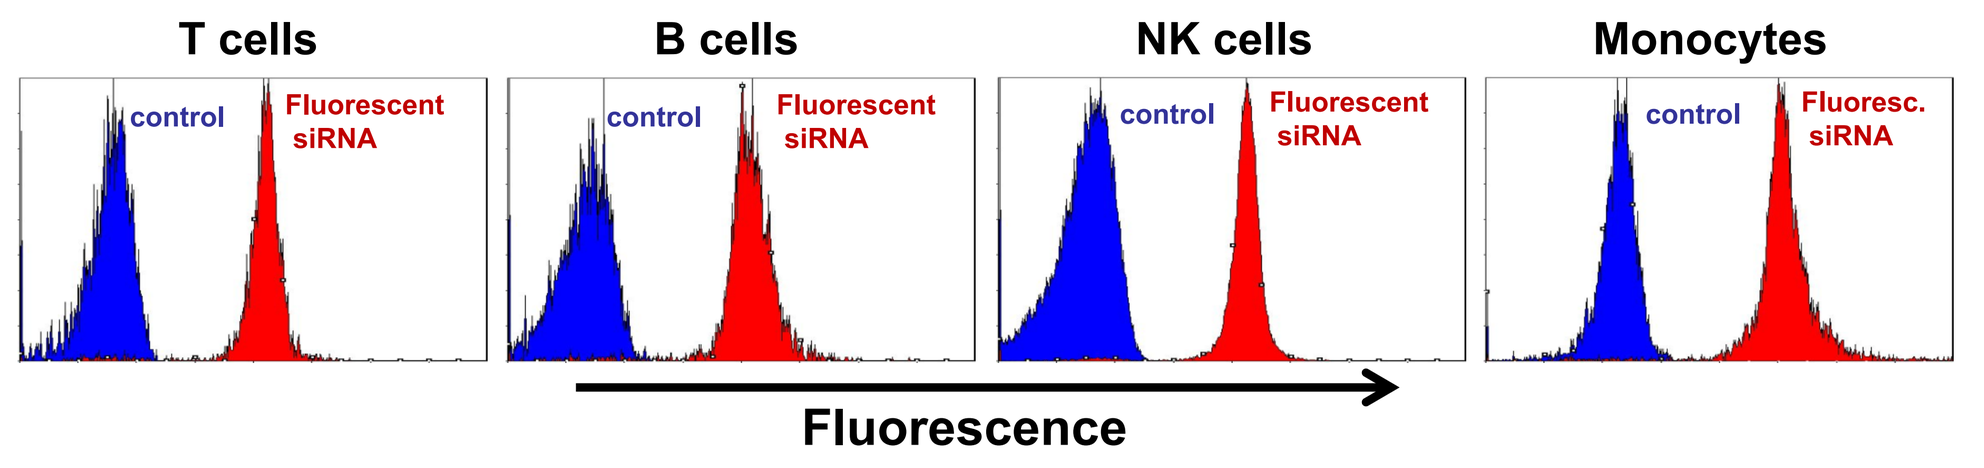
T cells
B cells
NK cells
Monocytes
control
Fluorescent siRNA
Fluoresc. siRNA
Fluorescence

Figure 5. PBMCs were transfected with fluorescently labeled siRNA and were stained for CD3, CD56 and CD19. Gated cell populations are shown as overlays of control (blue) vs. fluorescent siRNA (red).

This protocol enables efficient silencing of cbl-b in human T, B, NK cells and monocytes (Fig. 6, only T and NK cells are shown).

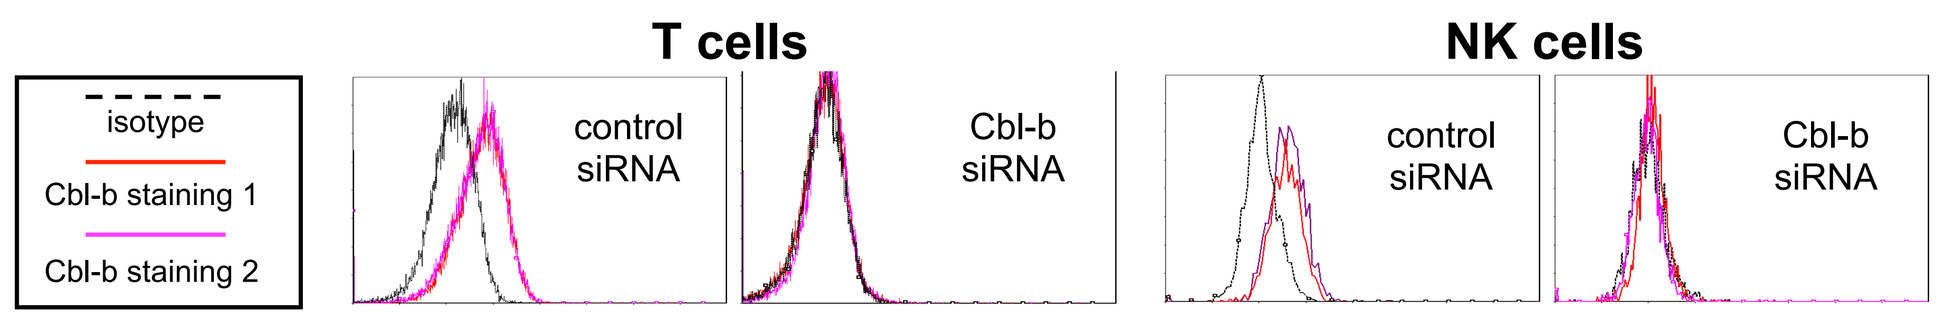
isotype
Cbl-b staining 1
Cbl-b staining 2
T cells
NK cells
control siRNA
Cbl-b siRNA

Figure 6. PBMCs were transfected with cbl-b or control siRNA and 2 days later stained for cbl-b, CD3 and CD56. Gated cell populations are shown as overlays.

Cbl-b silenced PBMCs respond to stimulation with enhanced anti-tumor cytokine production and tumor cell killing (Fig. 7).

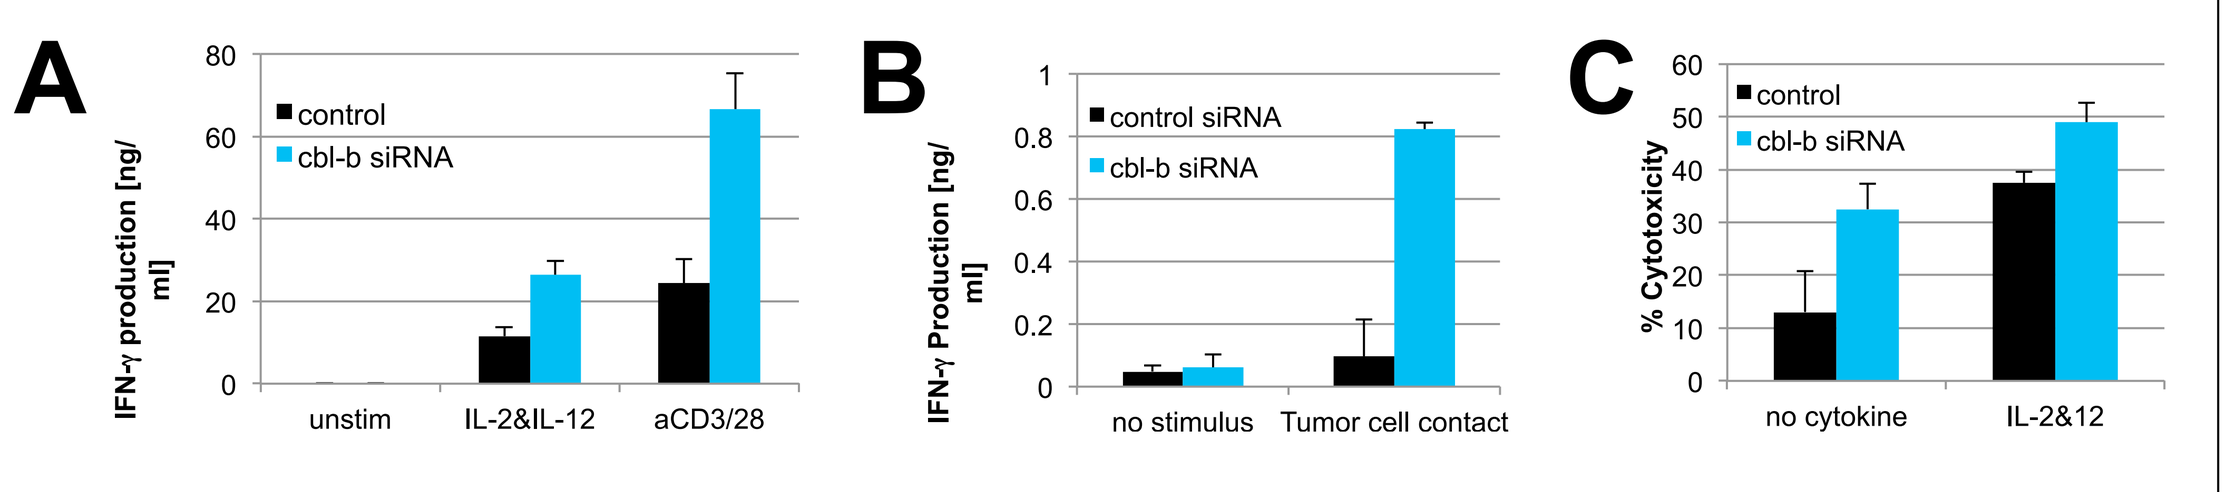
A
IFN-γ production [ng/ml]
control
cbl-b siRNA
unstim
IL-2&IL-12
aCD3/28
B
IFN-γ Production [ng/ml]
control siRNA
cbl-b siRNA
no stimulus
Tumor cell contact
C
% Cytotoxicity
control
cbl-b siRNA
no cytokine
IL-2&12

Figure 7. PBMCs were transfected with cbl-b or control siRNA. **A)** Cells were stimulated with rhu IL-2 & IL-12 or with anti-CD3/28 mAbs or in **B)** with irradiated K562 tumor cells and IFN-γ production was measured by ELISA. **C)** SKBR3 tumor cells were incubated for 4h with PBMCs either in the presence or absence of IL-2 and IL-12. Cytotoxicity was determined by LDH release of SKBR3 tumor cells.

Screening for an optimized cbl-b siRNA

A panel of siRNAs was synthesized using a proprietary algorithm (Biospring, Frankfurt). The efficiency of cbl-b silencing was determined by measuring cbl-b levels and induction of cytokines (Fig. 8). Overall, strong correlation of cbl-b silencing (both on mRNA and protein levels) to cytokine induction (IFN-γ, IL-2, TNF-α) was observed .

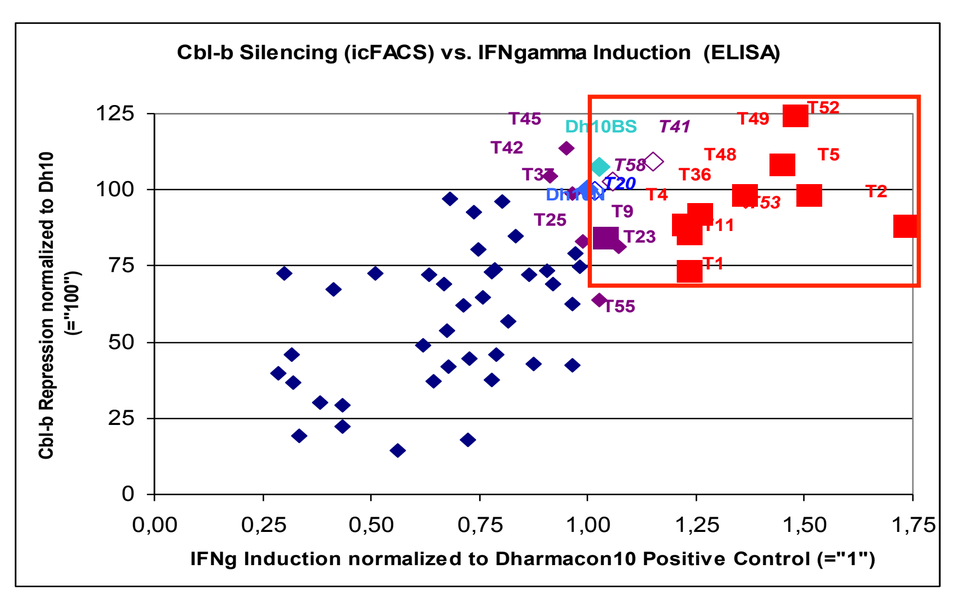
Cbl-b Silencing (icFACS) vs. IFNgamma Induction (ELISA)
Cbl-b Repression normalized to Dh10 (="100")
IFNg Induction normalized to Dharmacon10 Positive Control (="1")

Figure 8. PBMCs were transfected with different siRNAs and stimulated. Cbl-b silencing was related to control/isotype staining. IFN-γ levels were measured by ELISA and normalized to the induction ratio of the positive control Dh10. Transfections were performed in duplicates and in 2 different donors, results were averaged.

All siRNAs showing superior (red symbols) or similar (violet) cbl-b silencing compared to the positive control were resynthesized and retested. All selected siRNA candidates were confirmed to silence cbl-b and increase IFN-γ and IL-2 production (Fig. 9, only IL-2 production is shown). A further modified siRNA from the first screening round, T36OMe, was also included in the panel.

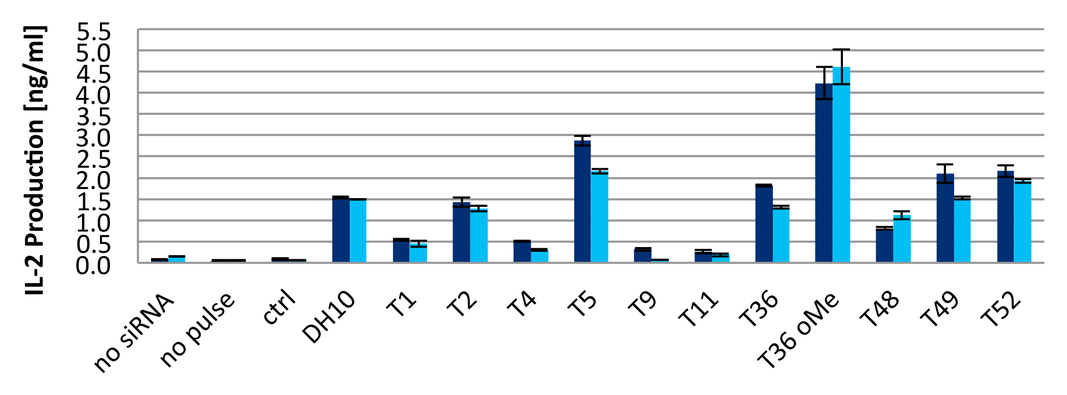
IL-2 Production [ng/ml]
no siRNA
no pulse
ctrl
DH10
T1
T2
T4
T5
T9
T11
T36
T36 oMe
T48
T49
T52

Figure 9. PBMCs were transfected with siRNAs (duplicates in light and dark blue) and IL-2 levels were determined.

An optimized siRNA for clinical application should be able to suppress cbl-b for at least several days. Fig. 10 shows that even in strongly proliferating cells cbl-b levels could be suppressed for more than 7 days. Concordantly, siRNA T36OMe mediated sustained production of IL-2 over several days.

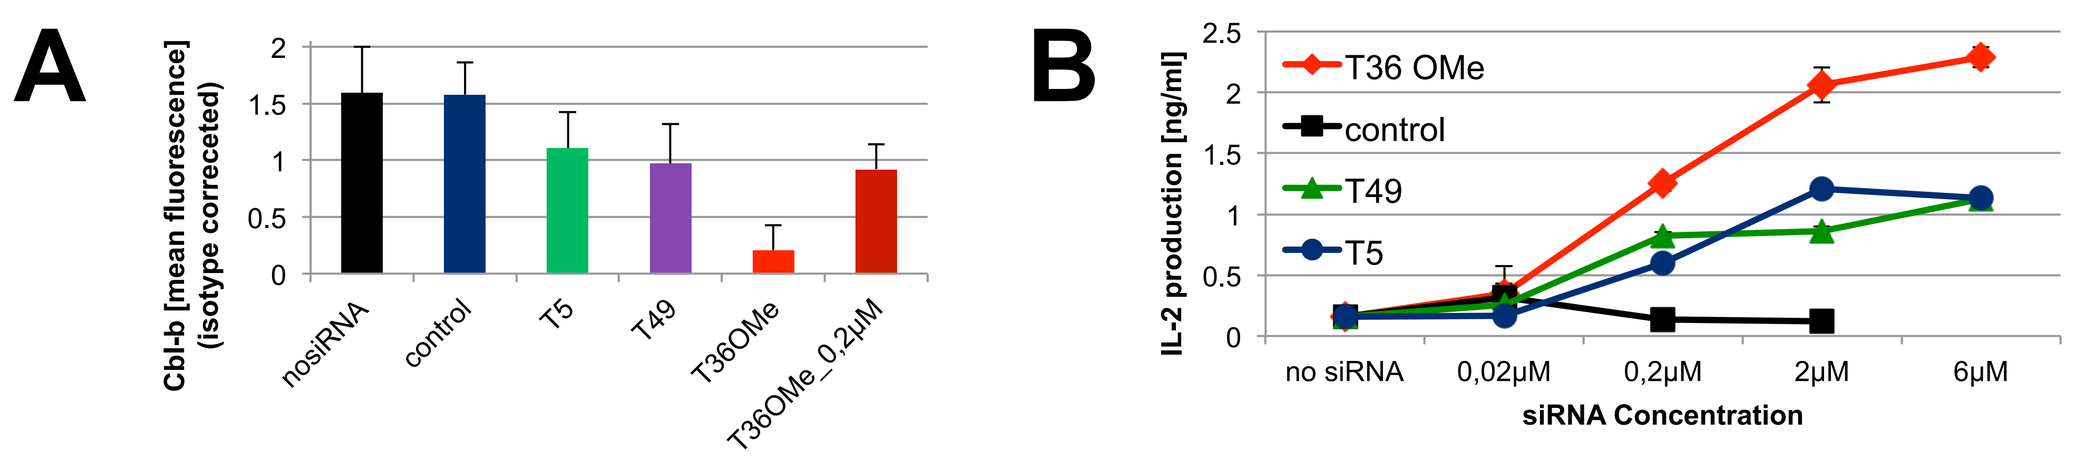
A
Cbl-b [mean fluorescence] (isotype corrected)
nosiRNA
control
T5
T49
T36OMe
T36OMe_0,2µM
B
IL-2 production [ng/ml]
T36 OMe
control
T49
T5
no siRNA
0,02µM
0,2µM
2µM
6µM
siRNA Concentration

Figure 10. A) PBMCs were CFSE labelled, transfected with siRNAs (used at 2µM unless indicated otherwise) and stimulated with antiCD3&28 mAbs for 7 days. Cells were gated on at least 3 completed cell divisions, and cbl-b levels of two independent transfections were determined by icFACS and averaged. **B**) PBMCs were transfected with indicated concentrations of siRNA, stimulated for 3 days and IL-2 production was measured by ELISA.

Based on these results, T36OMe siRNA was finally selected as optimized siRNA for therapeutic application of cbl-b Silencing.

Targeting cbl-b for cancer immunotherapy

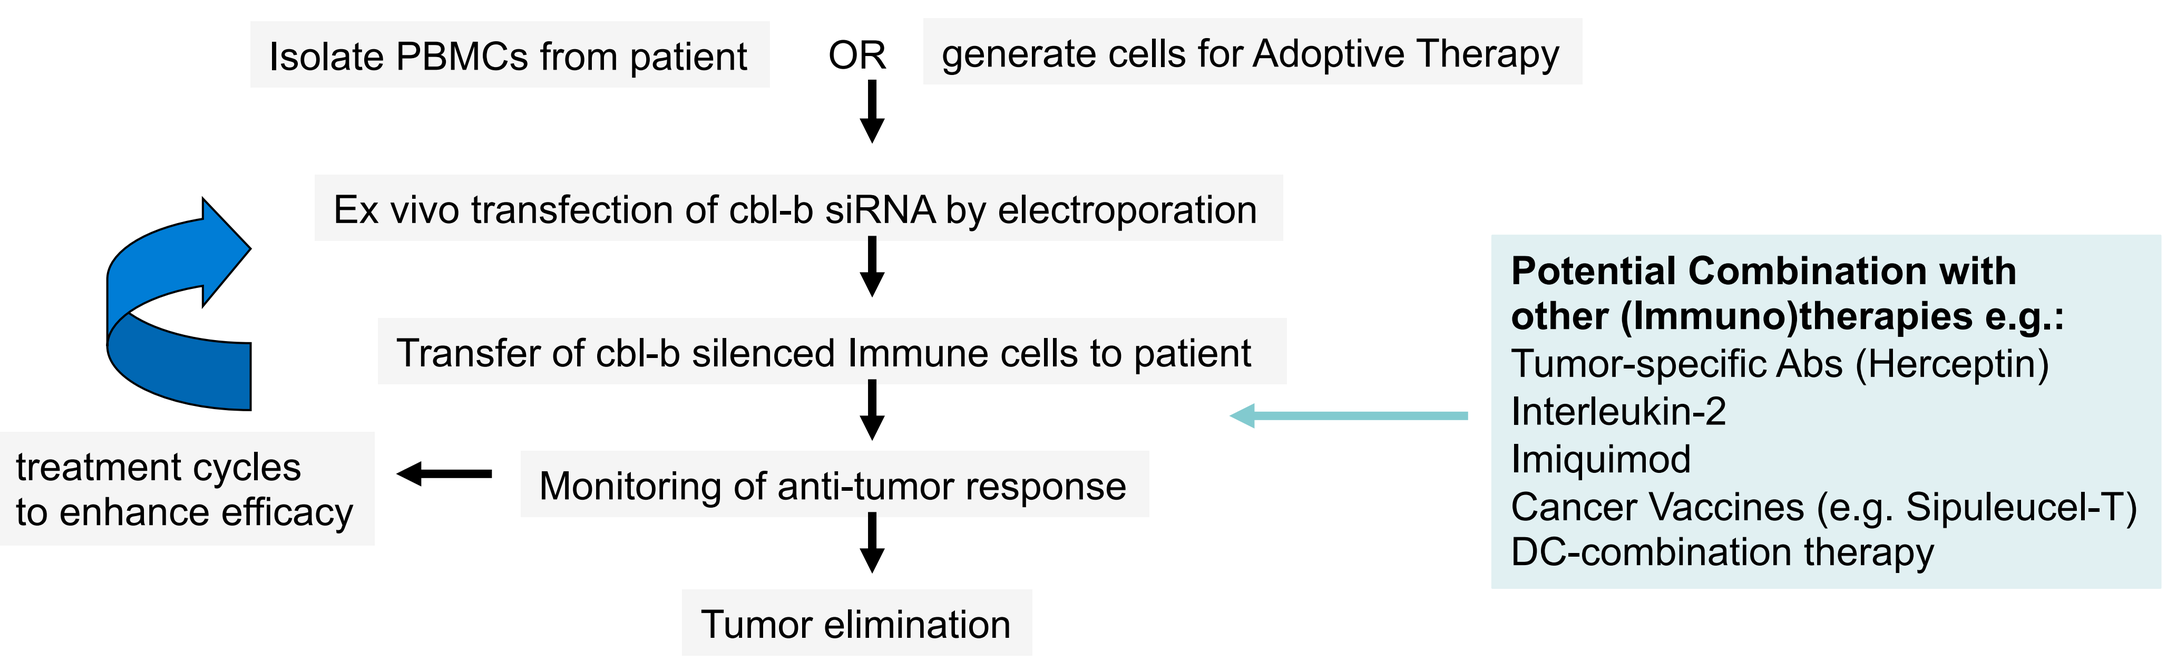
Isolate PBMCs from patient
OR
generate cells for Adoptive Therapy
Ex vivo transfection of cbl-b siRNA by electroporation
Transfer of cbl-b silenced Immune cells to patient
Monitoring of anti-tumor response
treatment cycles to enhance efficacy
Tumor elimination
Potential Combination with other (Immuno)therapies e.g.:
Tumor-specific Abs (Herceptin)
Interleukin-2
Imiquimod
Cancer Vaccines (e.g. Sipuleucel-T)
DC-combination therapy

Figure 11. Schematic outline of ex-vivo cbl-b silencing therapy either as a single therapy or in combination with ACT or other immunotherapies

Ex vivo cbl-b Silencing as adjuvant co-therapy for anti-tumor DC vaccination protocol

The murine tumor model data yielded a rationale for combining DC vaccination with cbl-b ex vivo silencing.
Hence, we aimed to enhance the anti-tumor immune reactivity of a patient with inoperable lung metastases (originating from a primary pancreatic tumor) undergoing a DC vaccination therapy with lysates of pancreatic tumor serving as antigen source.
A manufacture process was established for isolation, ex vivo silencing and re-administration of cbl-b silenced PBMCs to the patient (Fig. 12)

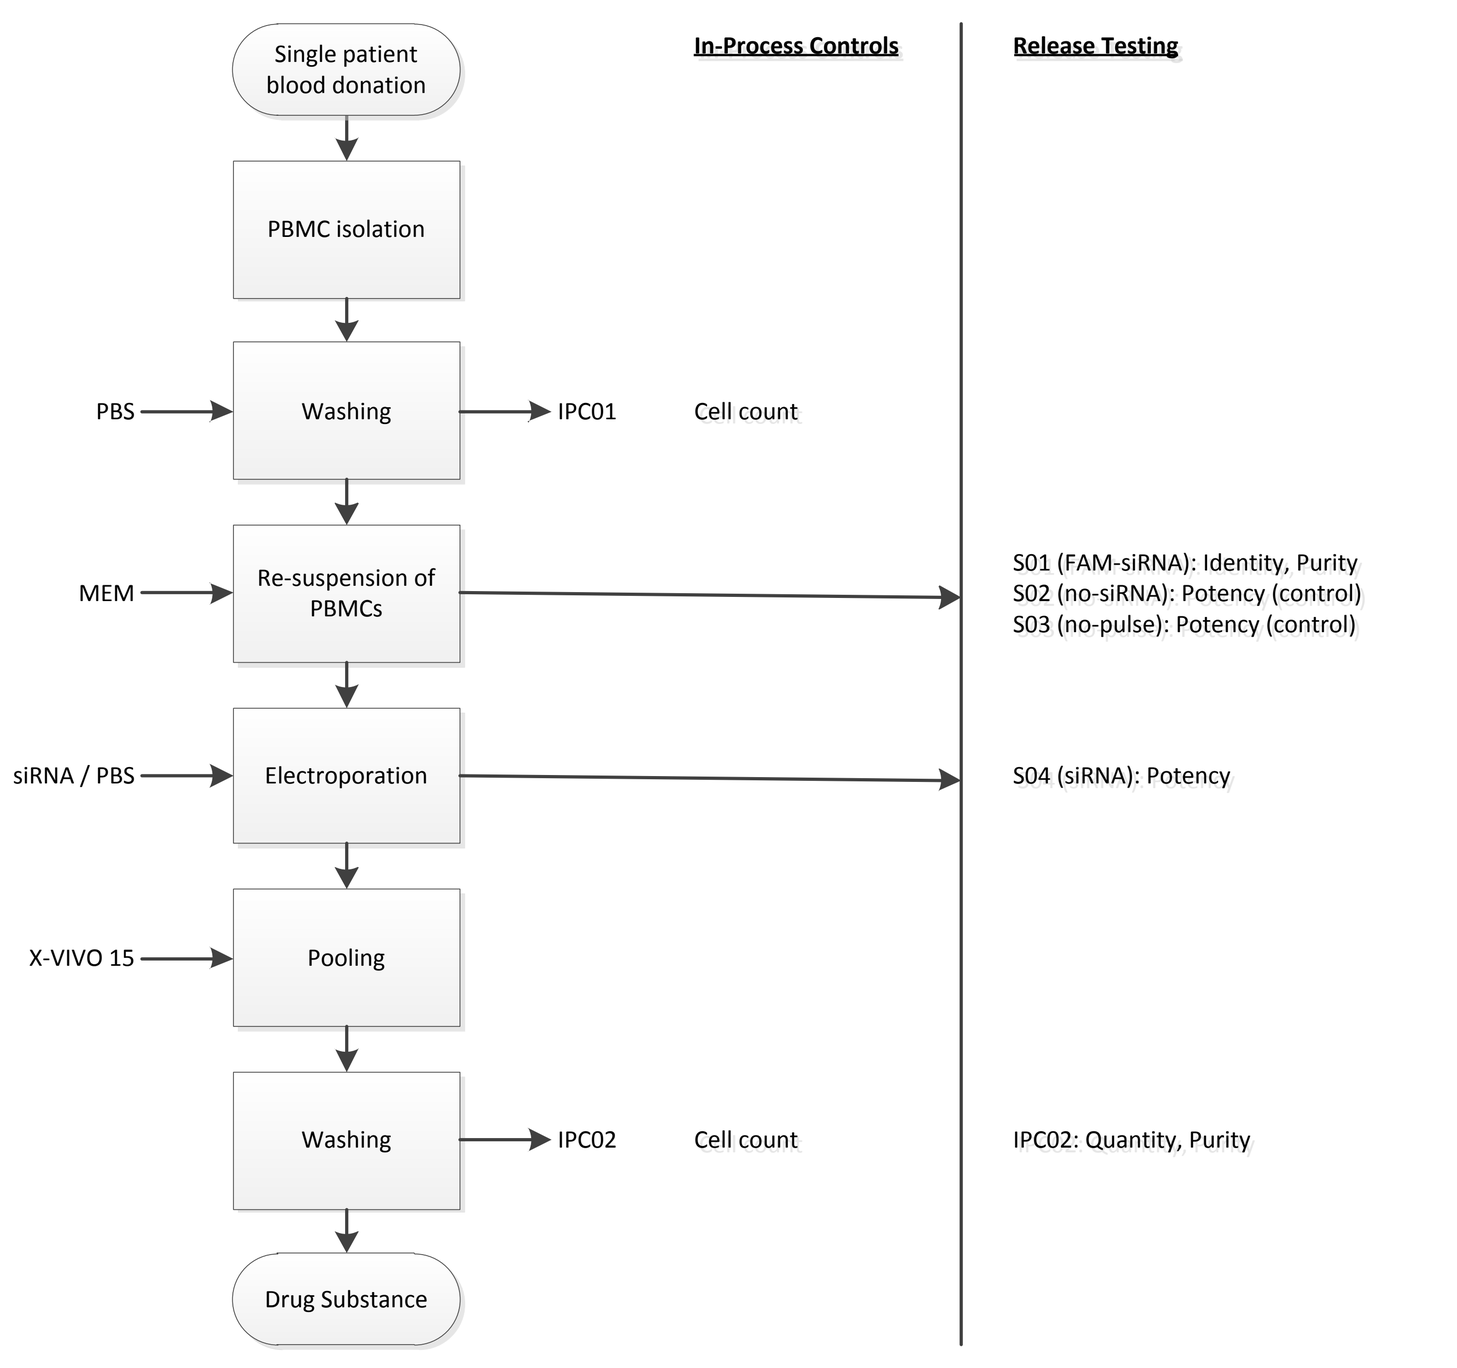
In-Process Controls
Release Testing
Single patient blood donation
PBMC isolation
PBS
Washing
IPC01
Cell count
MEM
Re-suspension of PBMCs
S01 (FAM-siRNA): Identity, Purity
S02 (no-siRNA): Potency (control)
S03 (no-pulse): Potency (control)
siRNA / PBS
Electroporation
S04 (siRNA): Potency
X-VIVO 15
Pooling
Washing
IPC02
Cell count
IPC02: Quantity, Purity
Drug Substance

Figure 12. Flowchart for the manufacture process of cbl-b silenced autologous patient PBMCs. In process controls and release testing was optimized to enable timely re-administration of cbl-b silenced PBMCs to the patient without any disruptive storage (e.g. freeze/thaw) procedures together with the antigen-loaded and activated DCs.

10 vaccination cycles were performed with intranodal co-administration of cbl-b silenced PBMCs together with antigen-loaded activated DCs. Prior to DC vaccination, a pre-dose of cbl-b silenced PBMCs was administrated without any DCs. The treatment was well tolerated without any severe adverse effects, and no side effects were noted when only cbl-b silenced PBMCs were transferred.

A

	Cbl-b silenced PBMCs [Mill. Cells]
Pre-Dose	58,5
Vacc. 1	55,5
Vacc. 2	130,8
Vacc. 3	112,9
Vacc. 4	72,4
Vacc. 5	43,7
Vacc. 6	46,8
Vacc. 7	46,4
Vacc. 8	33,6
Vacc. 9	32,4
Vacc. 10	31,2

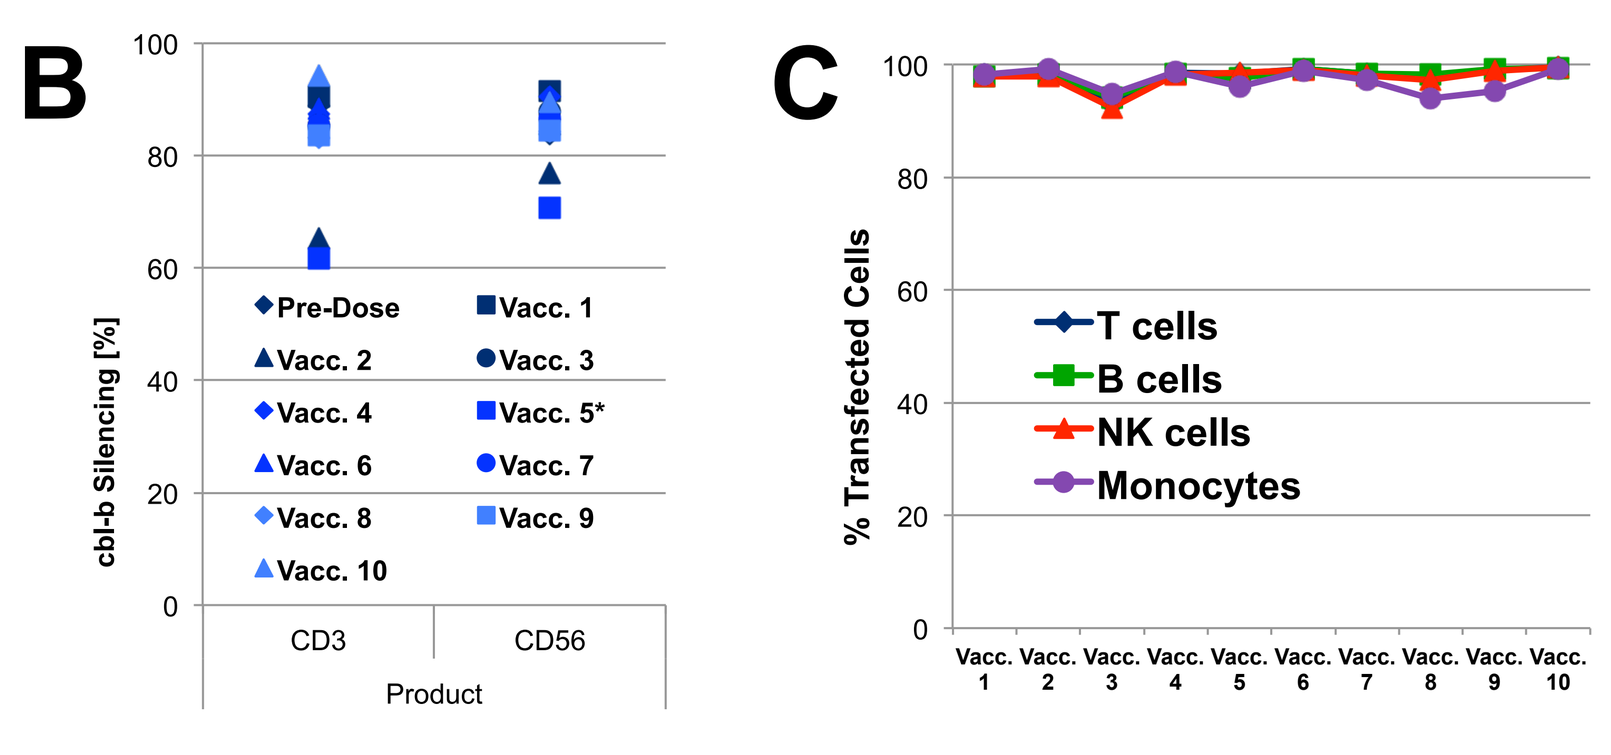
B
cbl-b Silencing [%]
Pre-Dose
Vacc. 1
Vacc. 2
Vacc. 3
Vacc. 4
Vacc. 5*
Vacc. 6
Vacc. 7
Vacc. 8
Vacc. 9
Vacc. 10
CD3
CD56
Product
C
% Transfected Cells
T cells
B cells
NK cells
Monocytes

Figure 13. A) Table showing the number of cbl-b silenced PBMCs that were transferred to the patient for each single vaccination. **B)** The percentage of cbl-b silencing was determined by icFACS comparing cbl-b silenced PBMCs (Product) with in-process control S02 (no siRNA) **C)** Transfection efficiency was determined using in process control S01 (FAM siRNA)

Effects of ex vivo cbl-b Silencing as adjuvant co-therapy for anti-tumor DC vaccination

The main aspect of cbl-b silenced PBMCs for enhancing anti-tumor immune reactivity in vivo is expected to be mediated by increased T cell proliferation, cytokine production and costimulation effects on antigen-presenting cells, as observed in the murine tumor model studies. However, these effects could not be determined in a non-invasive manner in the patient, thus functional consequences of cbl-b silencing were confirmed in TCR stimulated quality control sample aliquots (in process control PC02) of the product (Fig. 14)

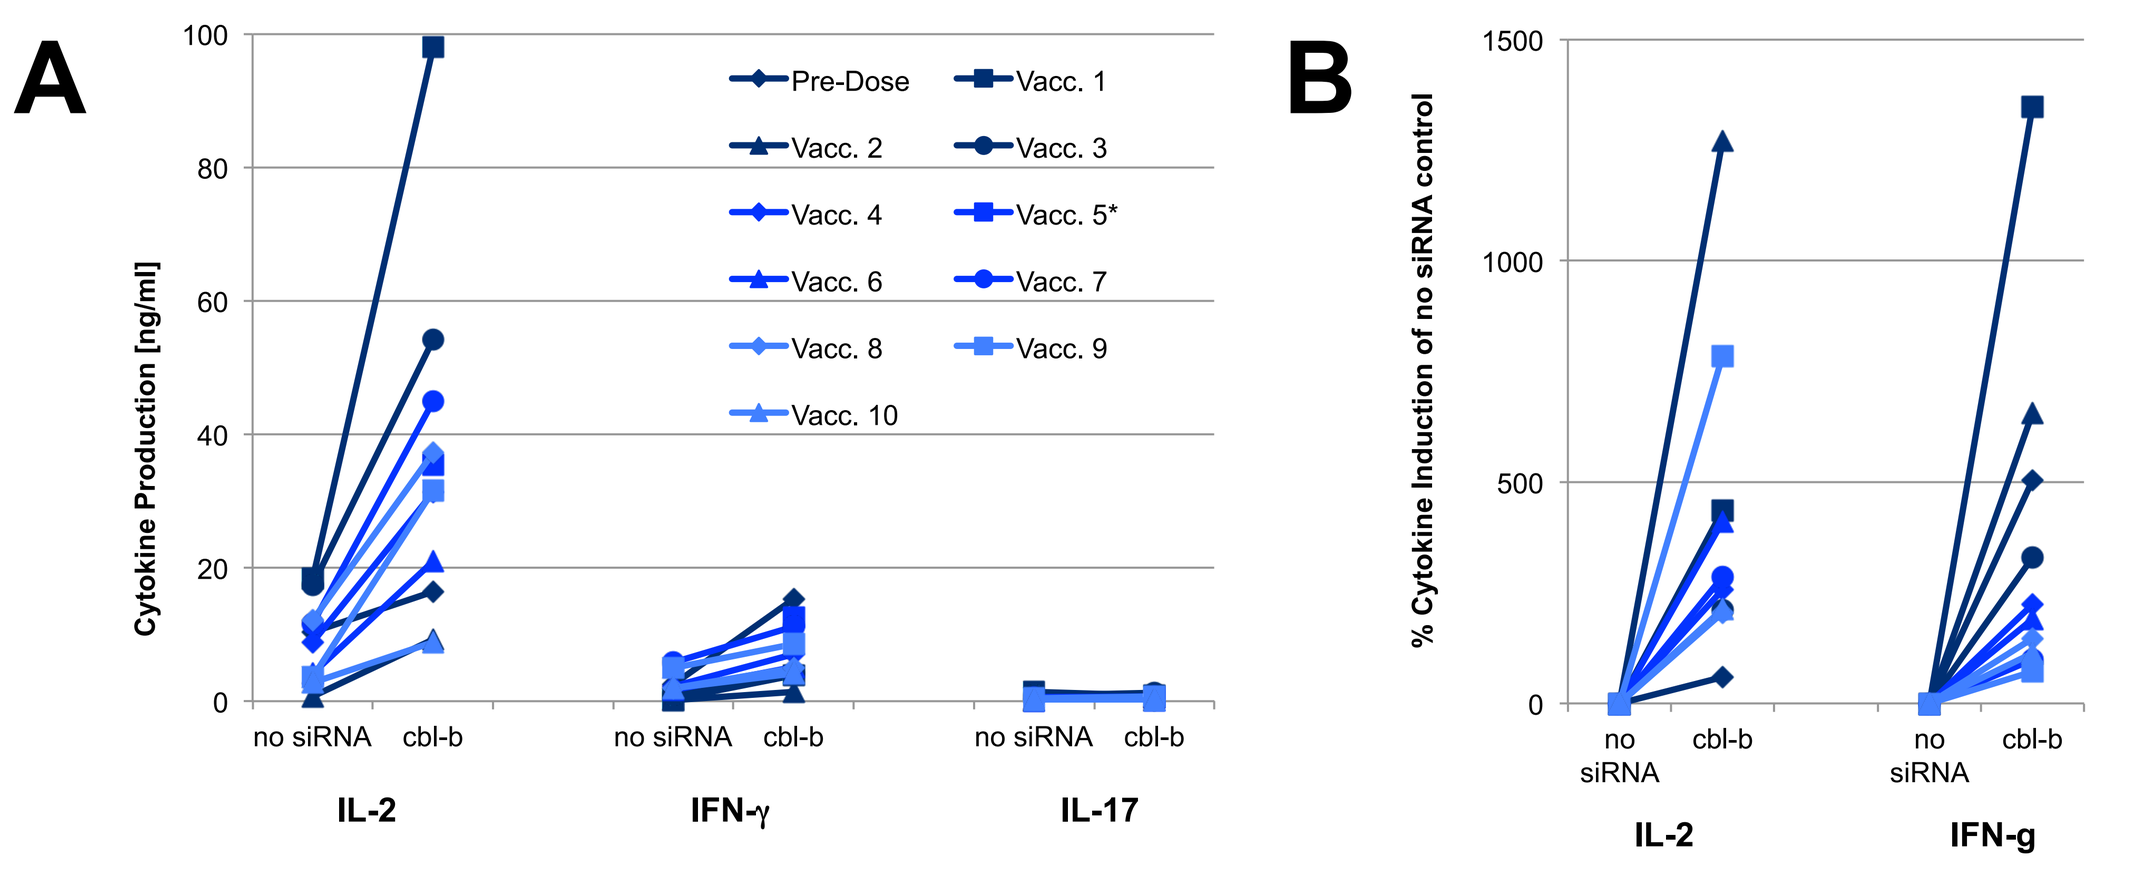
A
Cytokine Production [ng/ml]
Pre-Dose
Vacc. 1
Vacc. 2
Vacc. 3
Vacc. 4
Vacc. 5*
Vacc. 6
Vacc. 7
Vacc. 8
Vacc. 9
Vacc. 10
no siRNA
cbl-b
IL-2
IFN-γ
IL-17
B
% Cytokine Induction of no siRNA control
IL-2
IFN-g

Figure 14. A) Cytokine production of cbl-b silenced PBMCs and no siRNA control was determined by ELISA. **B)** Cytokine production of cbl-b silenced PBMCs is expressed as percent of no siRNA control

In addition, cbl-b silenced PBMCs were also tested for potential activation effects in the absence of external stimulation in vitro. No relevant cytokine production was observed without anti-CD3/28 stimulation in this set-up.

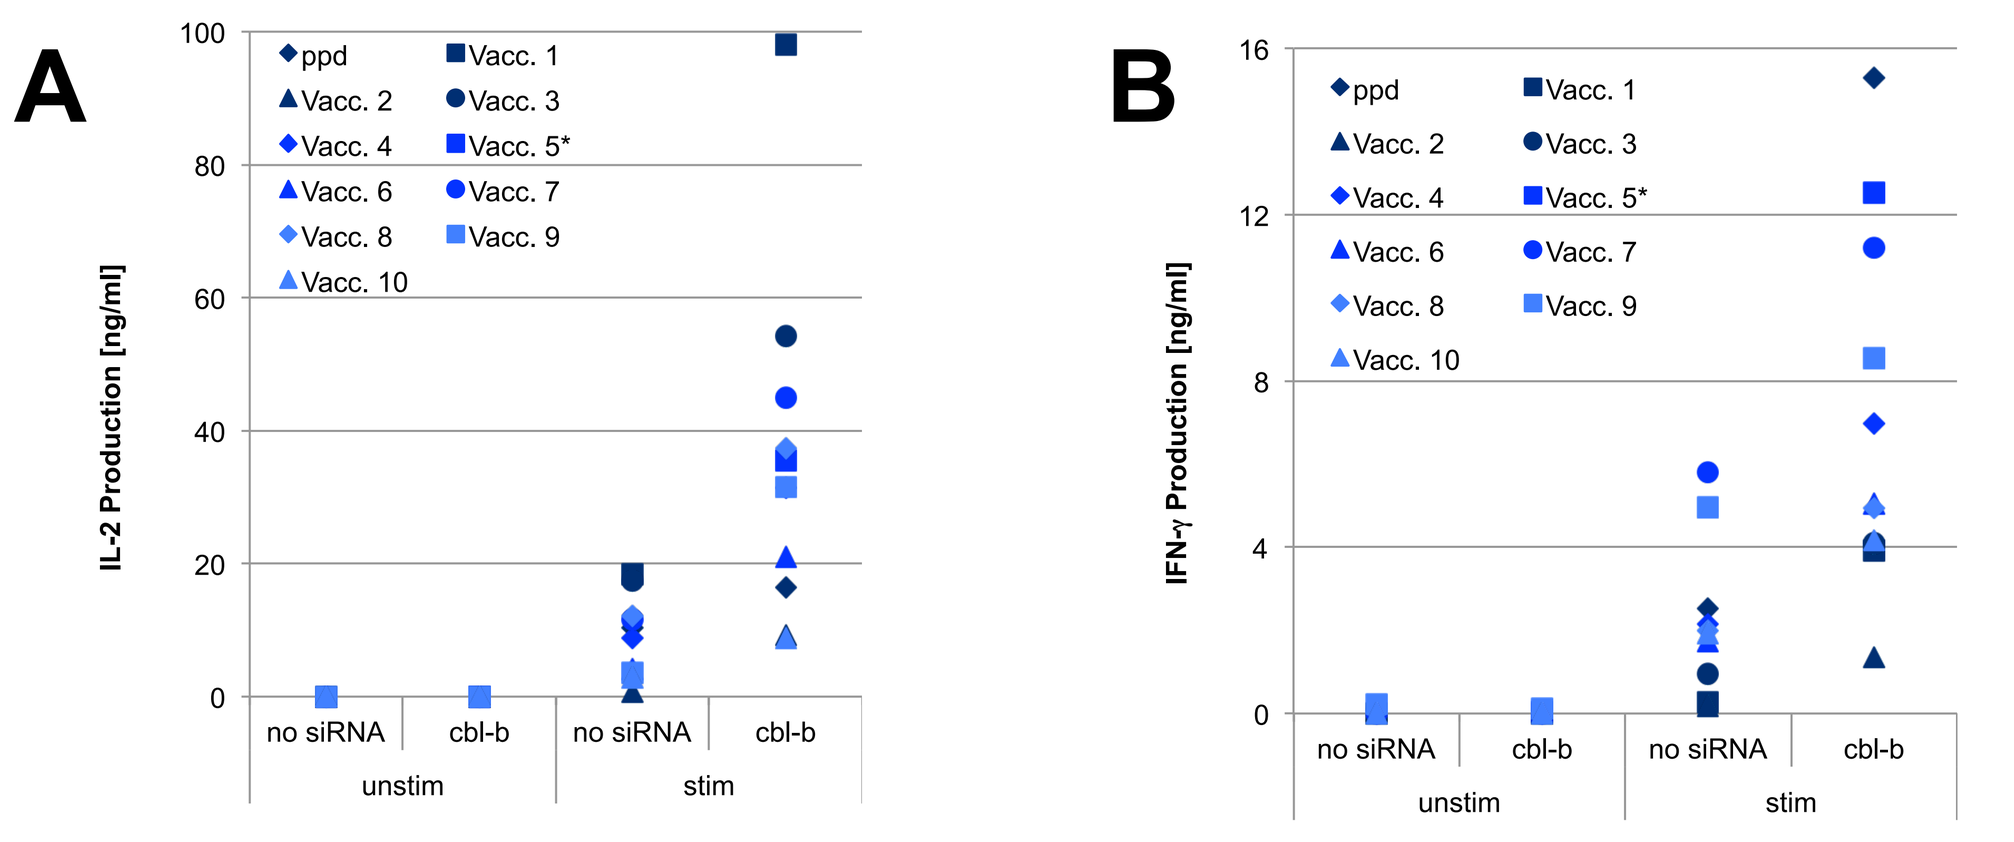
A
IL-2 Production [ng/ml]
B
IFN-γ Production [ng/ml]
ppd
Vacc. 1
Vacc. 2
Vacc. 3
Vacc. 4
Vacc. 5*
Vacc. 6
Vacc. 7
Vacc. 8
Vacc. 9
Vacc. 10
no siRNA
cbl-b
unstim
stim

Figure 15. A) & B) IL-2 and IFN-γ production of cbl-b silenced PBMCs and no siRNA control was determined by ELISA as above, either in the presence or absence of anti-CD3/28 stimulation

These results suggest, that ACT with ex-vivo cbl-b silenced T cells can have beneficial effects on anti-tumor immune activities, while maintaining a good safety and tolerability profile. The patient treated with this combined DC vaccination and cbl-b silencing therapy is currently in a treatment free period since 10 months (assessed as stable disease by MRT), with an overall survival since diagnosis of inoperable lung metastases of 19 months.

Phase I Study to assess Safety and Immunologic Activity of autologous cbl-b silenced PBMCs

A phase I clinical trial (open label) based on the adoptive infusion of autologous cbl-b silenced PBMC is being activated (Comprehensive Cancer Center Wake Forest University).
The objectives are to determine toxicities and maximum tolerated dose and immunologic effects (clinical response will also be documented).
Patients with refractory solid tumors will be enrolled into one of three sequential dose cohorts and receive an intravenous infusion of autologous cbl-b silenced PBMC every 14 days for a total of three infusions (up to a total of $6*10^9$).
Toxicity will be assessed using standard clinical and laboratory criteria. Blood lymphocyte function in response to antigenic stimulation and effects on regulatory, effector, and memory T cells will be determined.

Summary & Conclusions

Target Validation in murine Immune Cells

Enhanced activity of cbl-b deficient murine T cells:
- Increased production of cytokines with anti-tumor activity
- Increased proliferation of cbl-b deficient T cells
- TGF-β resistance

Absence of cbl-b enhances anti-tumor immunity
- Cbl-b knock-out mice are protected from tumors
- Transfer of cbl-b deficient T cells eradicates tumors
- Transfer of cbl-b silenced T cells suppresses tumor growth

Solid target validation in murine in vivo models

Target Validation in human Immune Cells

Enhanced activity of cbl-b silenced T cells:
- Increased production of cytokines with anti-tumor activity
- Increased proliferation of cbl-b silenced T cells
- TGF-β resistance

Enhanced activity of cbl-b silenced PBMCs:
- Silencing procedure established for human PBMCs
- Cbl-b silenced PBMCs respond stronger to tumor cell contact
- Cbl-b silenced PBMCs react stronger in ADCC

Solid target validation in human immune cells

Therapeutic Application of Cbl-b Silencing

Proprietary siRNA for optimal cbl-b silencing generated
- Increased production of cytokines with proprietary siRNA
- Sustained suppression of cbl-b expression in proliferating T cells achieved for more than 7 days

Clinical process for therapeutic application established
- siRNA production as GMP-compliant process
- Protocol for efficient silencing with in-process controls designed
- Feasibility of protocol and good safety profile confirmed

Phase I study for cbl-b silencing therapy has been initiated